Mechanism of Action, Preclinical Efficacy, and Safety Evaluation of ACP-711, a Novel $GABA_A$ Subunit α_3 Selective Modulator

Dipak V. Amrutkar,[1] Karin Sandager-Nielsen,[1] Tino Dyhring,[1] Thomas Amos Jacobsen,[1] Janus S. Larsen,[1] Pierandrea Muglia,[1] Sanjeev Pathak,[2] Hank Lin,[3] Robert K. Hofbauer[2]

[1]Saniona A/S, Glostrup, Denmark; [2]Acadia Pharmaceuticals, Inc., Princeton, New Jersey, USA; [3]Acadia Pharmaceuticals, Inc., San Diego, California, USA. **Presenting Author:** Robert K. Hofbauer <rhofbauer@acadia-pharm.com>

Poster No. 012
Neighborhood 17
Abstract No. 1587

INTRODUCTION

- Essential tremor (ET) is one of the most common neurological diseases and movement disorders[1,2]
- Although the exact etiology of ET is currently unknown, dysfunctional γ-aminobutyric acid (GABA) signaling may play a role[3,4]
- Many GABAergic medications, as well as other treatments for ET including β-blockers, antiseizure medications, and benzodiazepines have significant adverse events[4,5]
- GABA type A receptors ($GABA_ARs$) generally contain 5 subunits with several possible combinations of subtypes[6,7]

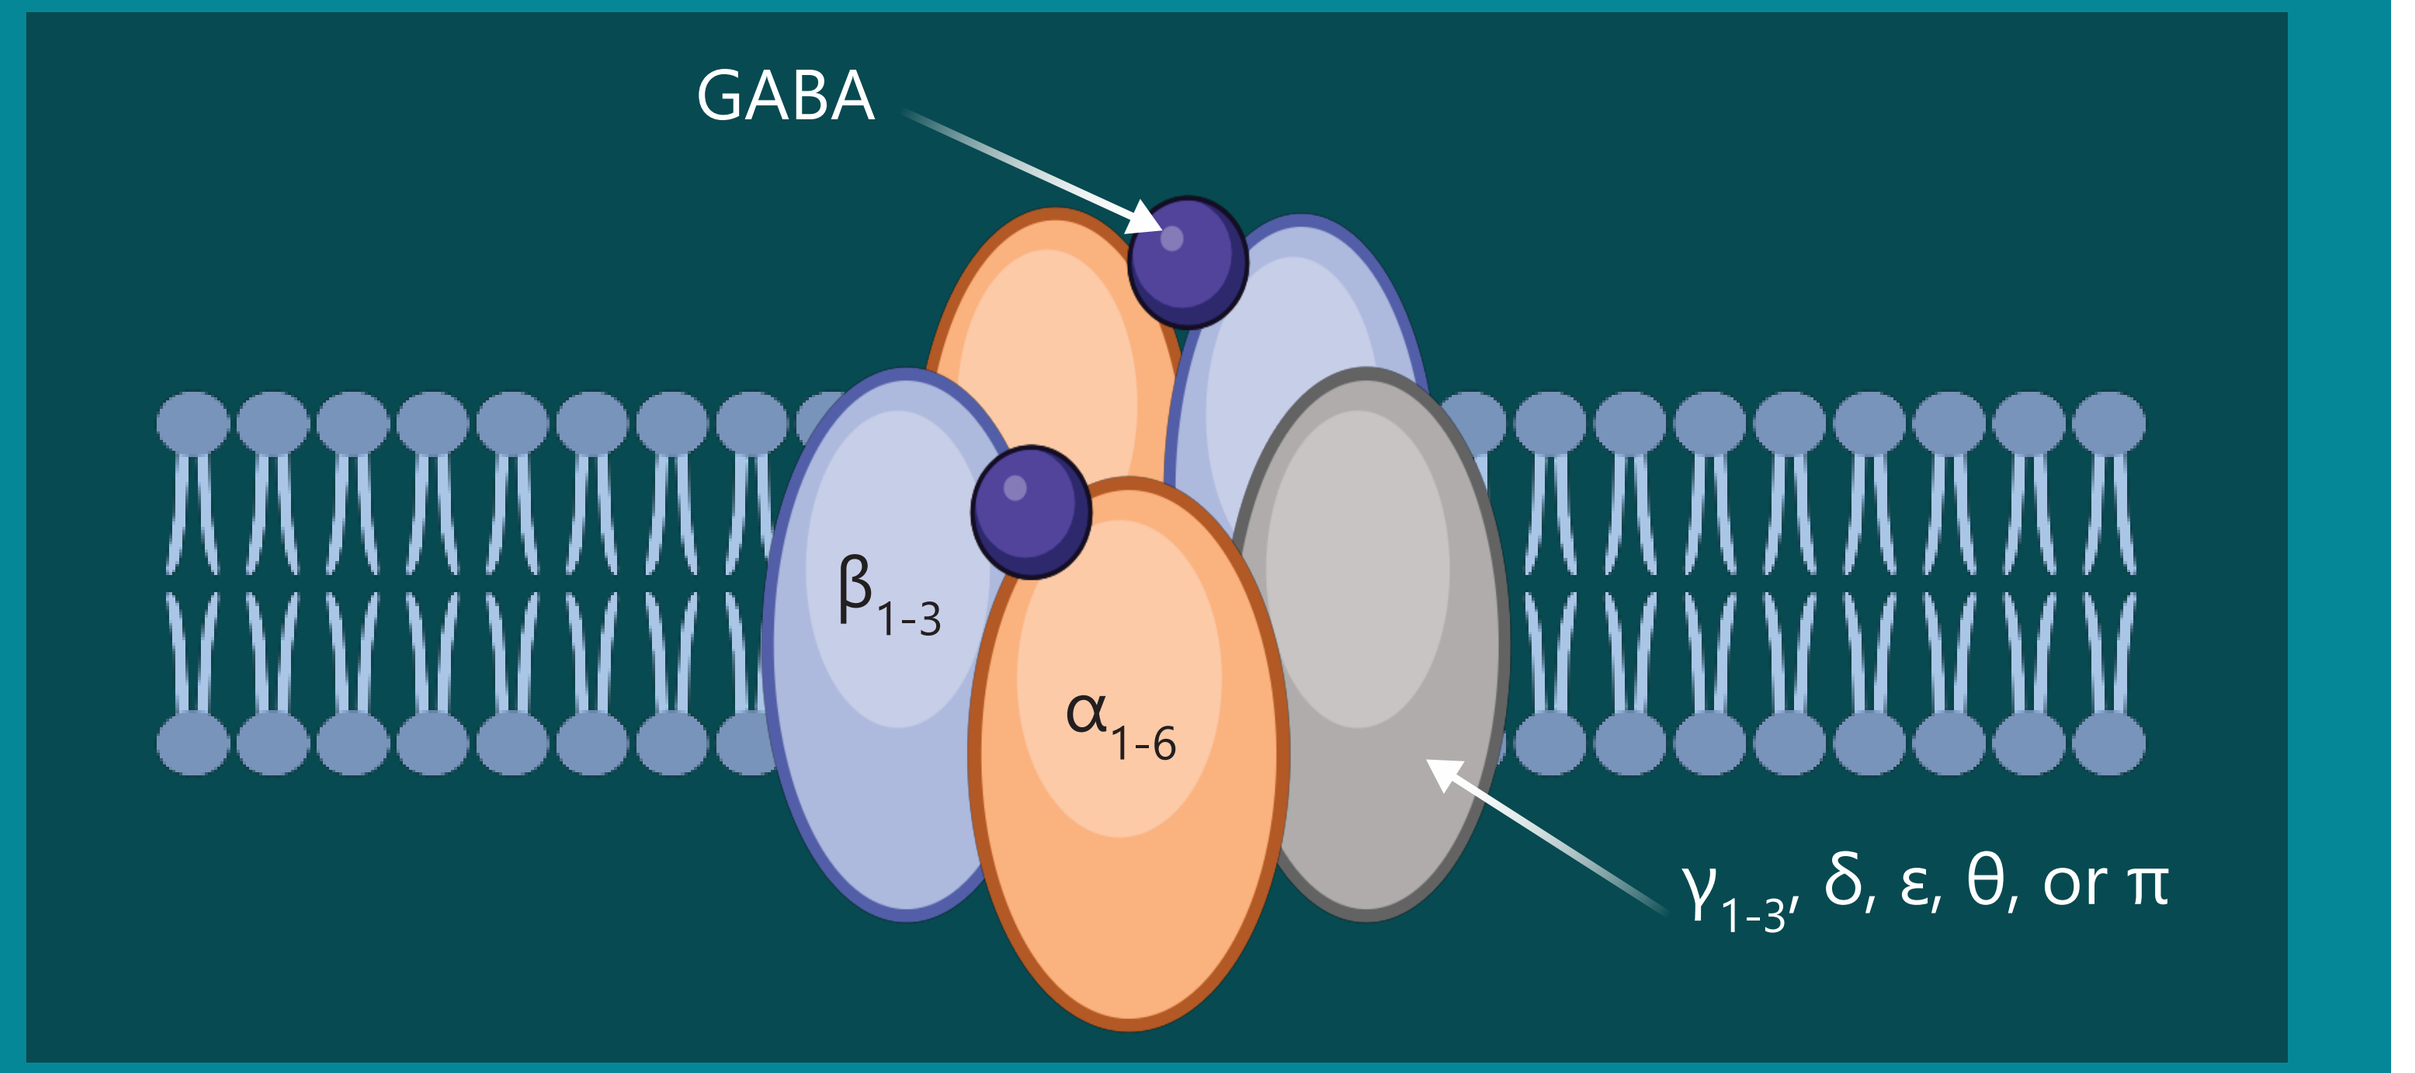

GABA
β_{1-3}
α_{1-6}
γ_{1-3}, δ, ε, θ, or π

- ACP-711 is a novel $GABA_AR$ subunit α_3–selective positive allosteric modulator (PAM) designed to have an improved safety profile compared with other GABAergic drugs
- Here, we evaluate the pharmacology, effectiveness, and safety of ACP-711 in vitro and in a preclinical animal model of ET

METHODS

- The efficiency of ACP-711 to modulate $GABA_AR$ subtype activity was explored in *Xenopus laevis* oocytes expressing human $GABA_ARs$ with subunit subtypes α_1, α_2, α_3, and α_5
- In vivo binding of ACP-711 to mouse forebrain $GABA_ARs$ was evaluated by administering injections of ^{3}H-flumazenil following oral ACP-711 doses
 - ^{3}H-flumazenil binds selectively and with high affinity to $GABA_ARs$[8]
- The antitremor properties of ACP-711 were assessed in rats using a harmaline model of ET
 - Propranolol was used as the positive control and vehicle as the negative control for treatment effect
- Potential safety concerns associated with $GABA_AR$ modulation were evaluated in rat models using nonselective $GABA_A$ PAM diazepam as a comparator
 - Sedative effects were tested by monitoring exploratory locomotor activity
 - Motor coordination was evaluated using a rotating accelerating rod (RotaRod) device

RESULTS

- ACP-711 was shown to be a potent PAM of $GABA_AR$ with an efficiency selectivity order of $\alpha_3\beta_2\gamma_2 > \alpha_2\beta_2\gamma_2 = \alpha_5\beta_2\gamma_2 >> \alpha_1\beta_2\gamma_2$ (**Figure 1**)

Figure 1. ACP-711 is a Potent Positive Allosteric Modulator of $GABA_AR$ (*Xenopus* Oocyte Model)

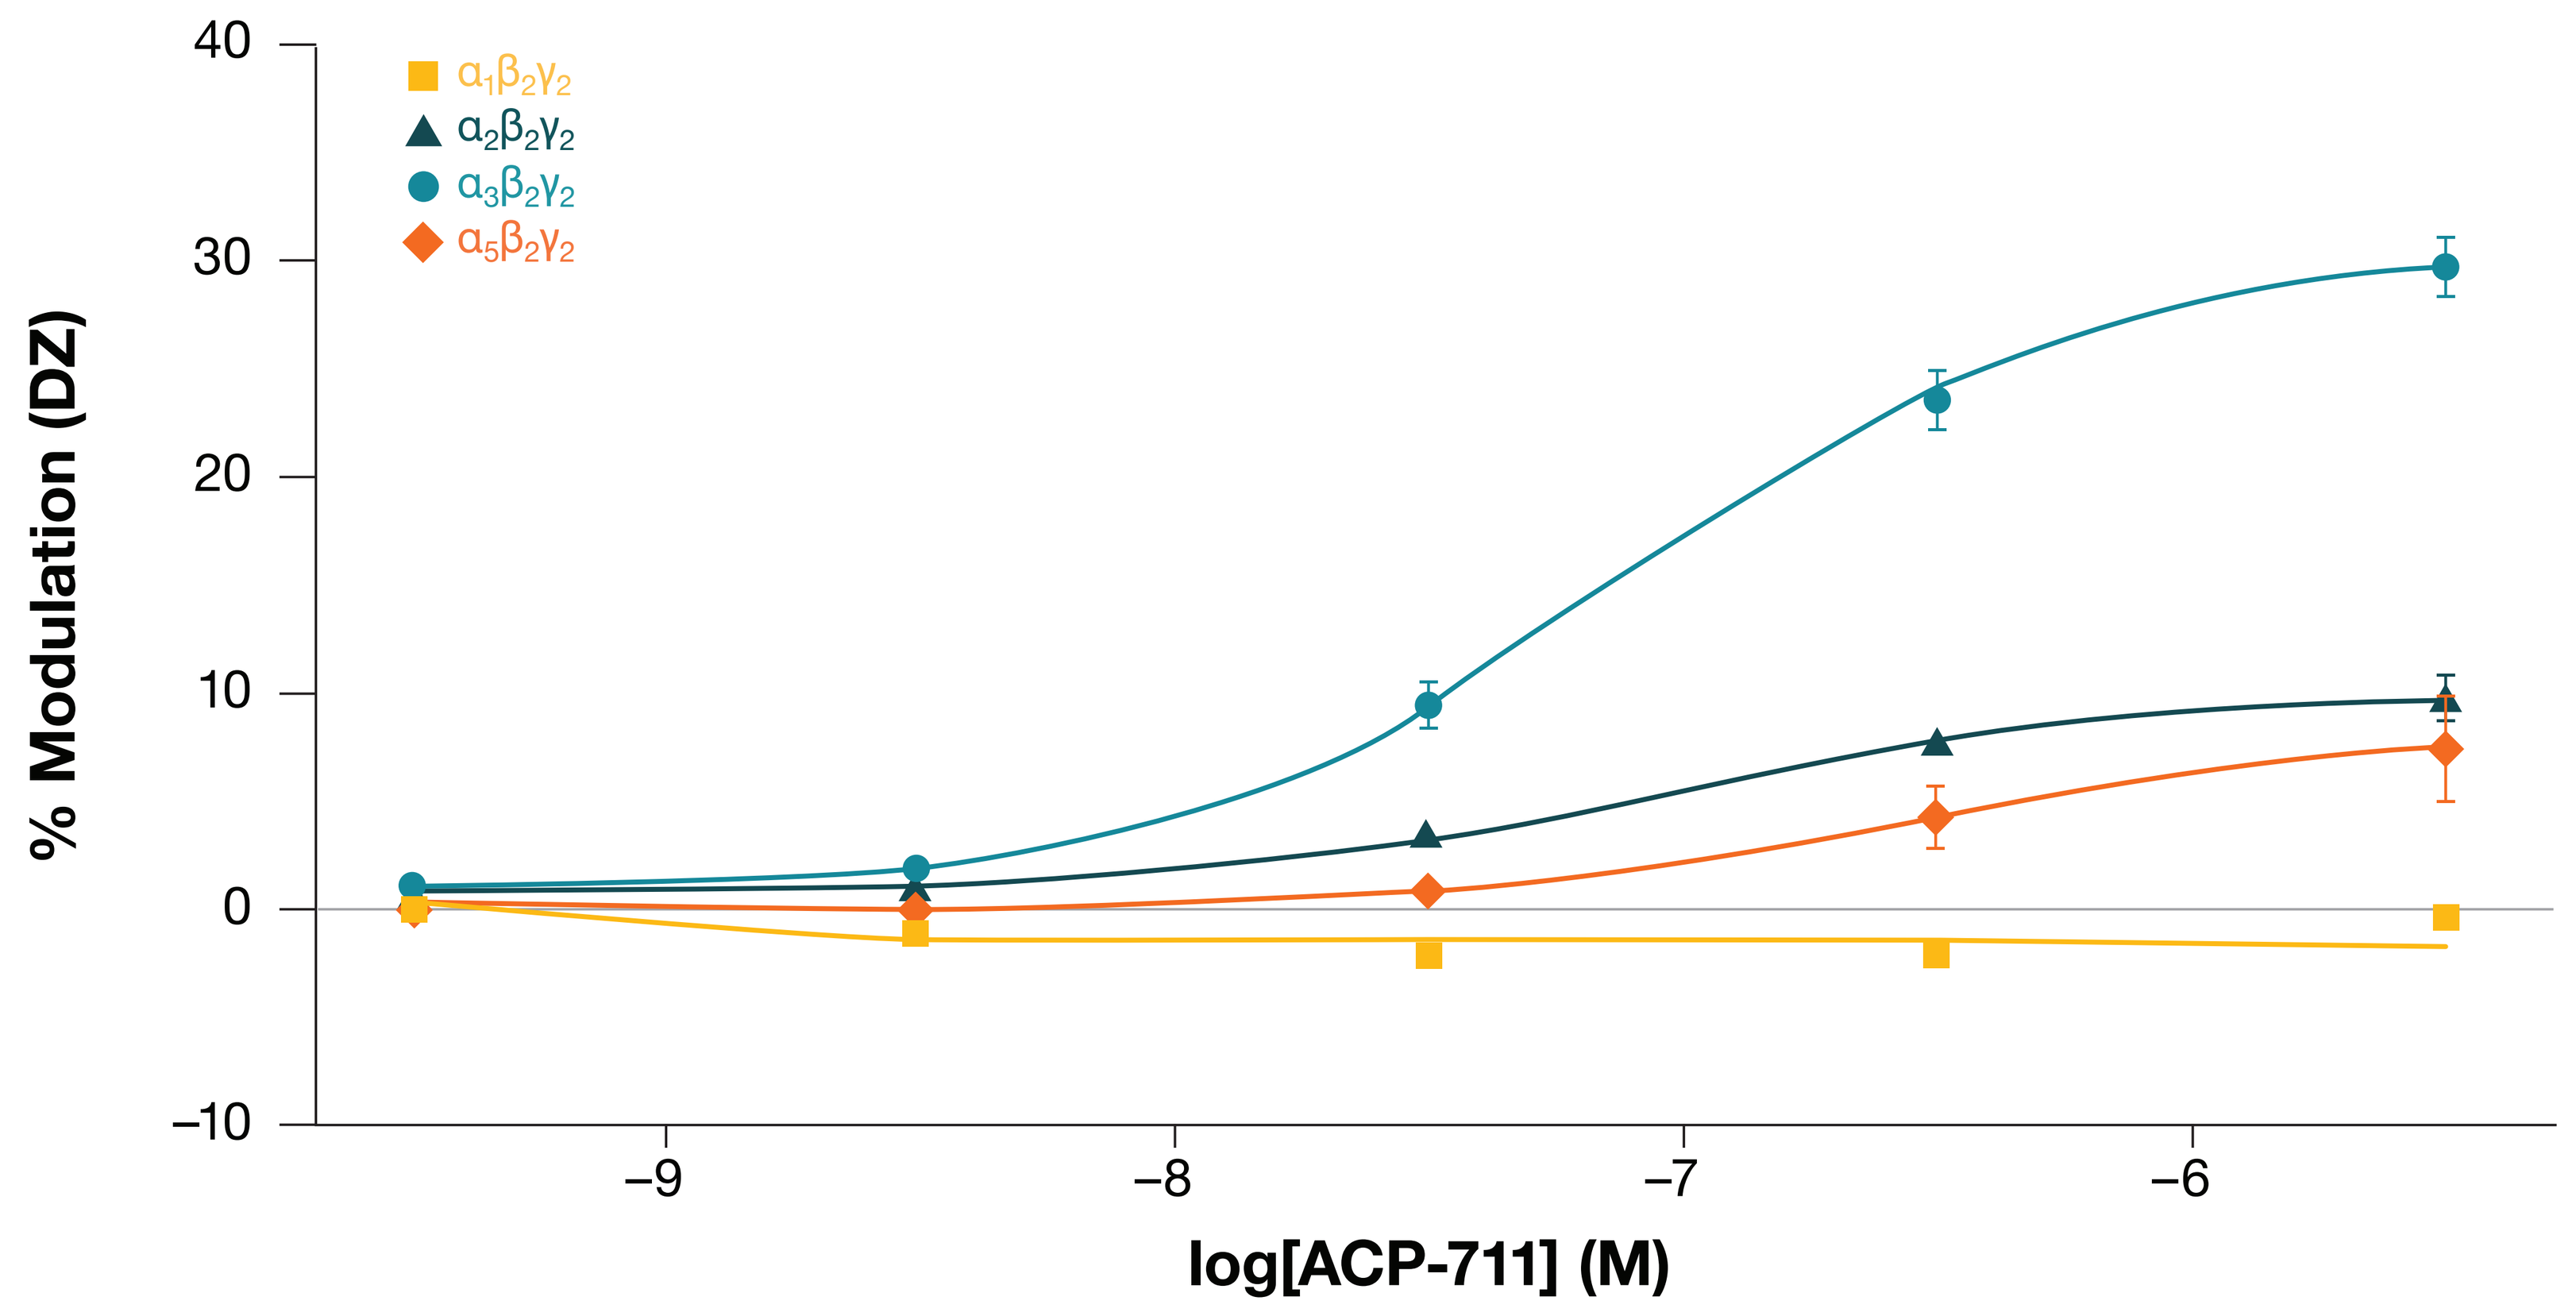

$\alpha_1\beta_2\gamma_2$
$\alpha_2\beta_2\gamma_2$
$\alpha_3\beta_2\gamma_2$
$\alpha_5\beta_2\gamma_2$
% Modulation (DZ)
40
30
20
10
0
–10
–9
–8
–7
–6
log[ACP-711] (M)

	$\alpha_1\beta_2\gamma_2$	$\alpha_2\beta_2\gamma_2$	$\alpha_3\beta_2\gamma_2$	$\alpha_5\beta_2\gamma_2$
E_{max}(%)	–1.4	10	31	8.5
EC_{50}(nM)	Undetermined	90	85	280
Number of oocytes	8	3	14	6

Values shown in the graph are means ± SE.
DZ, diazepam; EC_{50}, half-maximal effective concentration; E_{max}, maximum efficacy.

- Based on percentage inhibition of binding, the estimate of median effective dose (ED_{50}) ACP-711 was 0.7 mg/kg, corresponding to a plasma concentration of 0.32 μM (**Table 1**)

Table 1. Shows Adequate, Dose-Dependent Brain $GABA_A$ Receptor Occupancy (Mouse Forebrain Model)

Dose (mg/kg)[a]	Inhibition of binding (%)[b]	Plasma conc (μM)[c]
0.1	13	0.035
1	57	0.5
10	97	8.0

conc, concentration; $GABA_AR$, γ-aminobutyric acid type A receptor; LC-MS/MS, liquid chromatography-tandem mass spectrometry.
[a]Oral administration. [b]Percentage inhibition after 2 h oral administration of ACP-711 followed by tail vein injection of 2.0 μCi of ^{3}H-flumazenil 10 min before sacrifice and analysis of mouse forebrain tissue. [c]Determined using LC-MS/MS of plasma samples.

- ACP-711 dose-dependently decreased harmaline-induced tremors in rats, with a minimal effective dose of 1 mg/kg (**Figure 2**)

Figure 2. ACP-711 (Minimal Effective Dose) Was Associated With a Dose-Dependent Decrease in Harmaline-Induced Tremors (Rat Model)

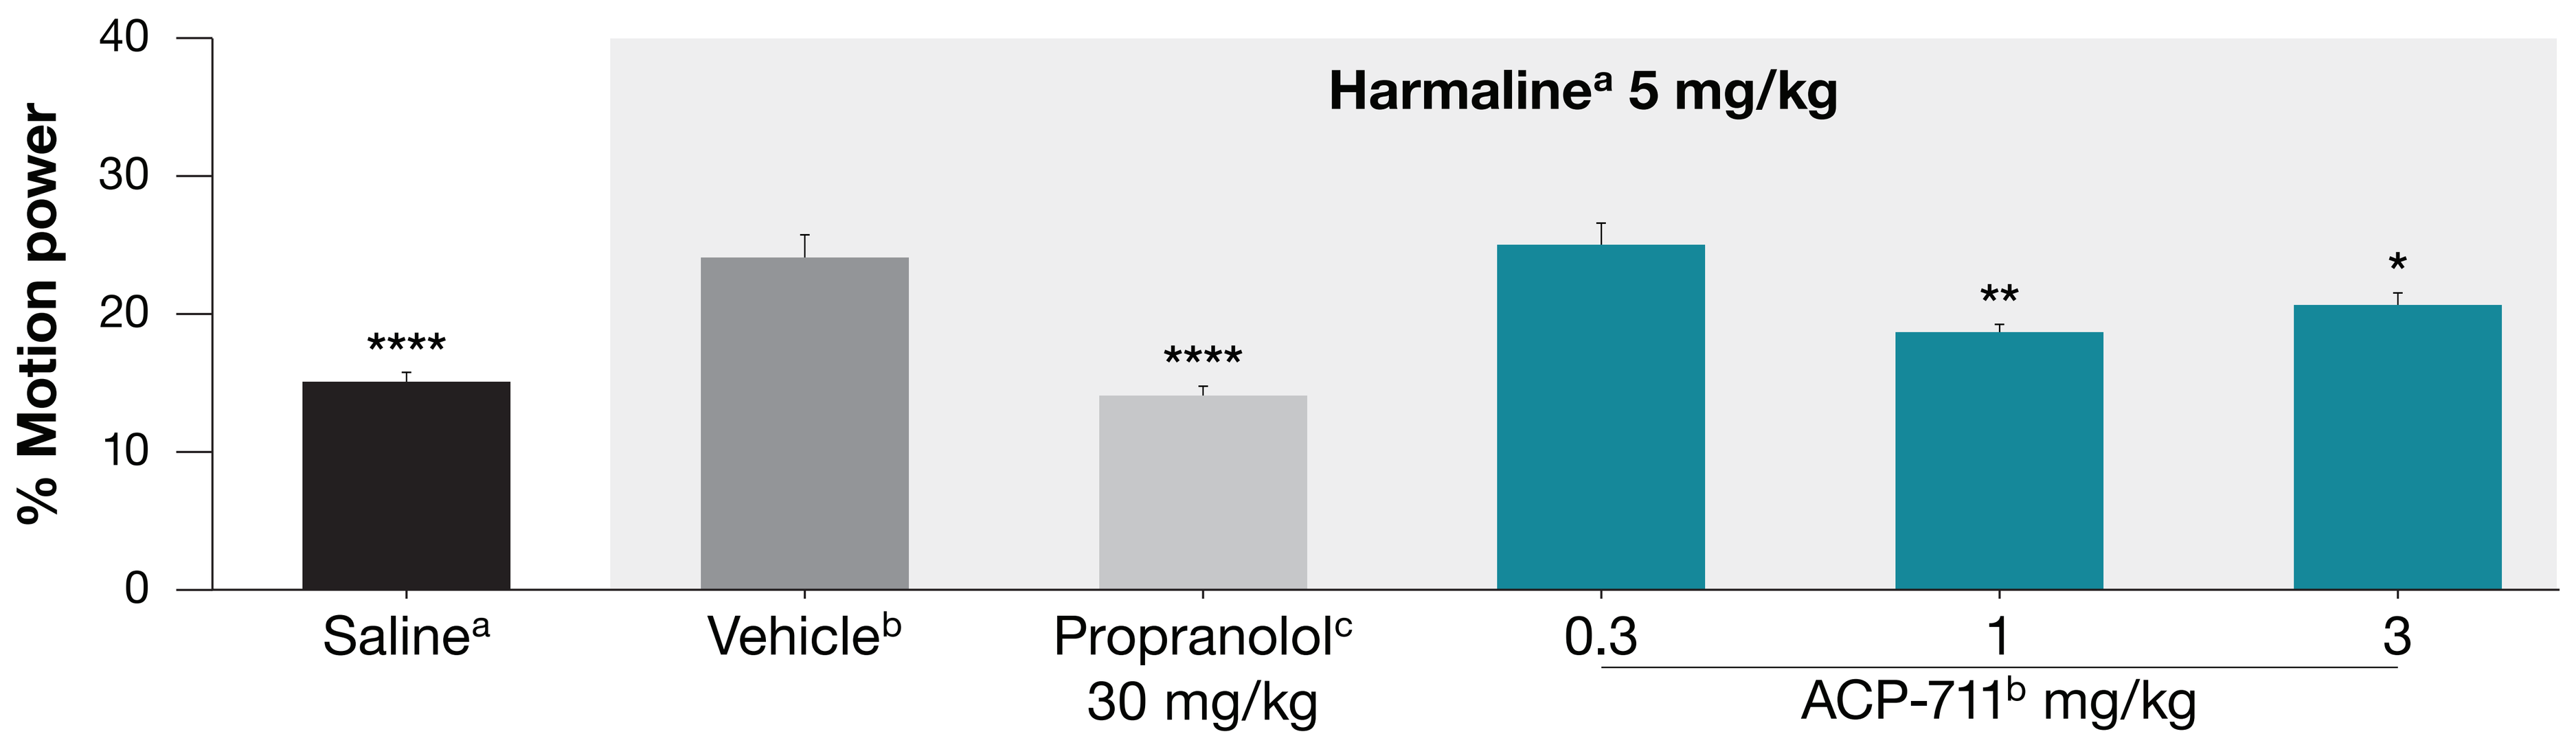

Harmaline[a] 5 mg/kg
% Motion power
40
30
20
10
0

**
*
Saline[a]
Vehicle[b]
Propranolol[c] 30 mg/kg
0.3
1
3
ACP-711[b] mg/kg

Values shown in the graph are means ± SEM. Motion power percentage is defined as the ratio of energy spent on tremor-like behavior divided by overall motion. A higher motion power percentage indicates worse tremors, with propranolol used as an active control for tremor reduction.
[a]Subcutaneous injection. [b]Oral administration. [c]Intraperitoneal injection.
* $P<0.05$; ** $P<0.01$; **** $P<0.0001$ vs vehicle.

- ACP-711 did not significantly affect exploratory motor activity or motor coordination, in contrast to diazepam (**Figure 3**)

Figure 3. ACP-711 Preserves Exploratory Behavior Without Sedative Effects Compared With Diazepam (Rat Model)

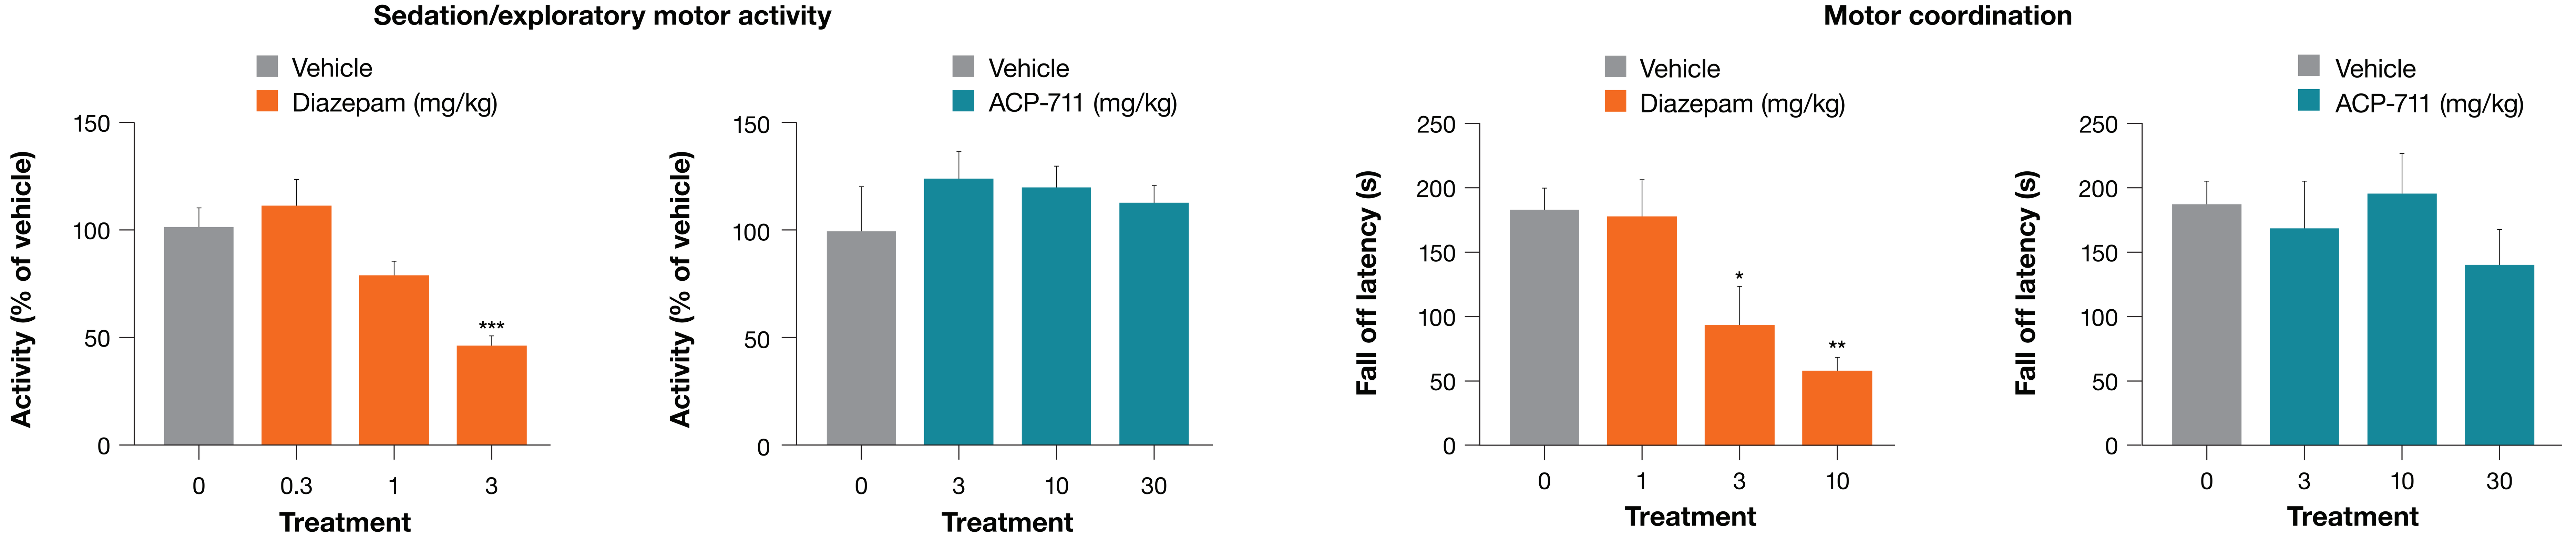

Sedation/exploratory motor activity
Motor coordination
Vehicle
Diazepam (mg/kg)
Activity (% of vehicle)
150
100
50
0

0
0.3
1
3
Treatment
Vehicle
ACP-711 (mg/kg)
Activity (% of vehicle)
0
3
10
30
Treatment
Vehicle
Diazepam (mg/kg)
Fall off latency (s)
250
200
150
100
50
0
*
**
0
1
3
10
Treatment
Vehicle
ACP-711 (mg/kg)
Fall off latency (s)
0
3
10
30
Treatment

Values shown in the graphs are means ± SEM. All substances administered orally. A decrease in exploratory motor activity relative to vehicle indicates sedative effects. A reduced time for rats to fall off the RotaRod (fall off latency) indicates reduced motor coordination.
* $P<0.05$; ** $P<0.01$; *** $P<0.001$ vs vehicle.

CONCLUSIONS

- ACP-711 is a novel selective modulator of $GABA_ARs$ containing the α_3 subunit
- In a rodent model, ACP-711 has shown effectiveness in treating harmaline-induced tremors with no detectable safety signals at doses up to 30 mg/kg
- ACP-711 demonstrates potential as a safe and effective treatment option for ET; further studies are warranted

REFERENCES

1. Sepulveda Soto MC, Fasano A. *Clin Park Relat Disord*. 2020;3:100031.
2. Song P, et al. *J Glob Health*. 2021;11:04028.
3. Kuo SH. *Neurotherapeutics*. 2023;20(2):372-374.
4. Bruno E, et al. *Cochrane Database Syst Rev*. 2016;10(10):CD009682.
5. Pal PK. *Ann Indian Acad Neurol*. 2011;14(suppl 1):S25-S28.
6. Jacob TC, et al. *Nat Rev Neurosci*. 2008;9(5):331-343.
7. Sallard E, et al. *Cell Mol Life Sci*. 2021;78(13):5341-5370.
8. Peris-Yague A, et al. *Eur Neuropsychopharmacol*. 2020;41:106-117.

DISCLOSURES

DVA, **KS-N**, **TD**, **TAJ**, **JSL**, and **PM** are employees of Saniona A/S and may own stock. **SP**, **HL**, and **RKH** are current or former employees of Acadia Pharmaceuticals Inc., and may own stock.

ACKNOWLEDGMENTS

Medical writing support was provided by Citrus Health Group, Inc. (Chicago, Illinois), and was funded by Acadia Pharmaceuticals Inc. (San Diego, California).

To receive a copy of this poster, scan the QR code via the barcode reader application. By requesting this content, you agree to receive a one-time communication using automated technology. Message and data rates may apply. Links are active for 30 days after the congress presentation.

Poster presented at the American Academy of Neurology (AAN) 2026 Annual Meeting; April 18-22, 2026; Chicago, Illinois, USA